UNIVERSITY of MARYLAND
SCHOOL OF PHARMACY
CENTER FOR TRANSLATIONAL MEDICINE

WOCKHARDT

Pharmacokinetics of WCK 2349 in Healthy Indian Male Adults under Fasting Conditions

Jing Niu[1], Vijay Ivaturi[1], Joga Gobburu[1], Rakesh Chugh[2], Ashima Bhatia[2]
[1]Center for Translational Medicine, School of Pharmacy, University of Maryland [2]Department of Clinical Research and Development, Wockhardt

LEARN & APPLY

Background & Objective

- The global spread of methicillin resistant *Staphylococcus aureus* (MRSA) in hospital and, more recently, in community is an unmet medical need[1,2]. WCK 2349, a novel fluoroquinolone, is being developed by Wockhardt as an oral anti- MRSA agent.

- The primary objective of this analysis was to characterize the pharmacokinetics (PK) of oral WCK 2349 in healthy adult Indian male volunteers under fasting conditions using Phase 1 data that included one single dose study with 7 dose levels (200 mg, 400 mg, 600 mg, 800 mg, 1000 mg, 1200 mg and 1500 mg) and one multiple dose study with 4 dose levels (600 mg, 800 mg, 1000 mg and 1200 mg twice daily (BID) for five days).

Methods

- Intensive venous blood samples were collected per protocol and WCK 2349 plasma concentrations were measured with LC-MS/MS.

- 1322 observations from 70 subjects (10 subjects per dose level) after single dose administration and 528 observations form 24 subjects (6 subjects per dose level) in steady state after multiple dose administration were analyzed using a non-compartmental approach with Phoenix WinNonlin.

Results

Table 1: Mean Pharmacokinetic Parameters of WCK 2349 for all dose levels

			$AUC_{(tau)}$ (ug·hr/mL)		$AUC_{(0\text{-}infinity)}$ (ug·hr/mL)		CL (L/hr)		Cmax (ug/mL)		$t_{1/2}$ (hr)		Vd (L)	
STUDY	DOSE (mg)	N	Mean	SD	Mean	SD	Mean	SD	Mean	SD	Mean	SD	Mean	SD
S.D.	200	10			31.58	5.92	6.51	1.07	5.11	1.31	6.12	1.26	57.08	14.26
S.D.	400	10			66.84	10.92	6.12	0.92	9.12	2.21	8.38	1.43	73.71	15.06
S.D.	600	10			81.48	21.67	7.84	2.04	12.16	1.57	7.67	1.45	85.93	25.31
S.D.	800	10			138.97	26.16	5.99	1.38	17.84	3.86	7.29	0.97	62.50	14.99
S.D.	1000	10			157.36	32.82	6.60	1.32	19.15	4.73	7.57	1.68	71.95	21.38
S.D.	1200	10			203.39	49.74	6.21	1.45	23.44	3.87	8.09	1.08	71.41	14.26
S.D.	1500	10			226.06	51.22	7.03	2.00	25.64	4.42	8.39	1.29	83.32	18.30
BID×5D	600	6	91.11	20.25	125.61	43.33	6.85	1.41	14.81	2.25	6.86	1.60	65.76	10.11
BID×5D	800	6	120.10	21.12	156.06	31.21	6.82	1.14	20.56	2.67	6.85	0.97	67.50	14.54
BID×5D	1000	6	186.36	38.10	254.65	65.24	5.56	1.16	29.68	4.09	7.46	1.11	59.15	12.18
BID×5D	1200	6	225.21	39.84	321.76	80.34	5.45	0.85	33.78	4.21	8.68	1.54	66.91	5.97

Figure 1: Mean WCK 2349 Concentration-Time Profiles in single (left panel) and multiple dose studies (right panel)

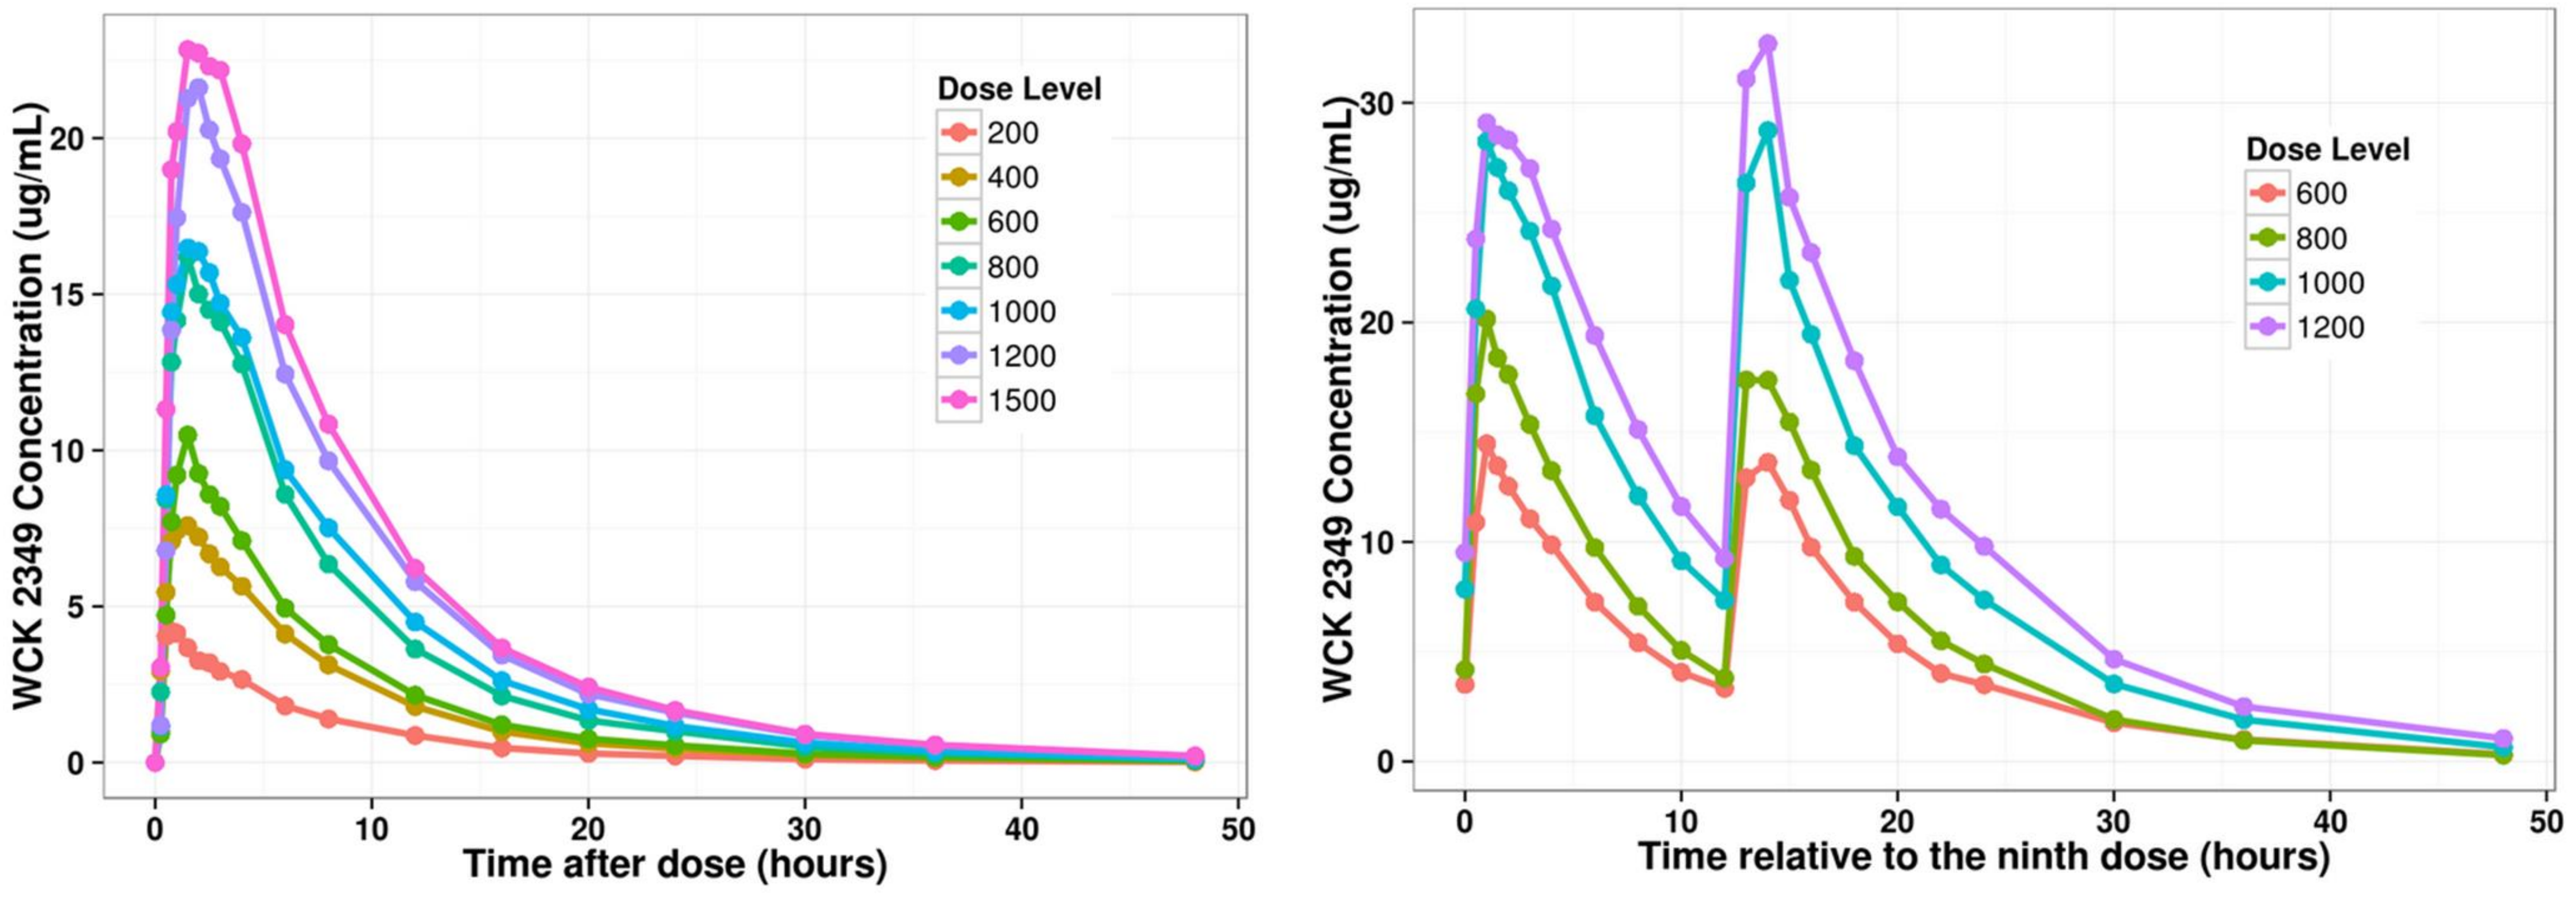
WCK 2349 Concentration (ug/mL)
Time after dose (hours)
Dose Level
200
400
600
800
1000
1200
1500
WCK 2349 Concentration (ug/mL)
Time relative to the ninth dose (hours)
Dose Level
600
800
1000
1200

Results/Discussion

Figure 2: Mean C_{max} (left panel) and $AUC_{(0\text{-}infinity)}$ (right panel) in single and multiple dose studies

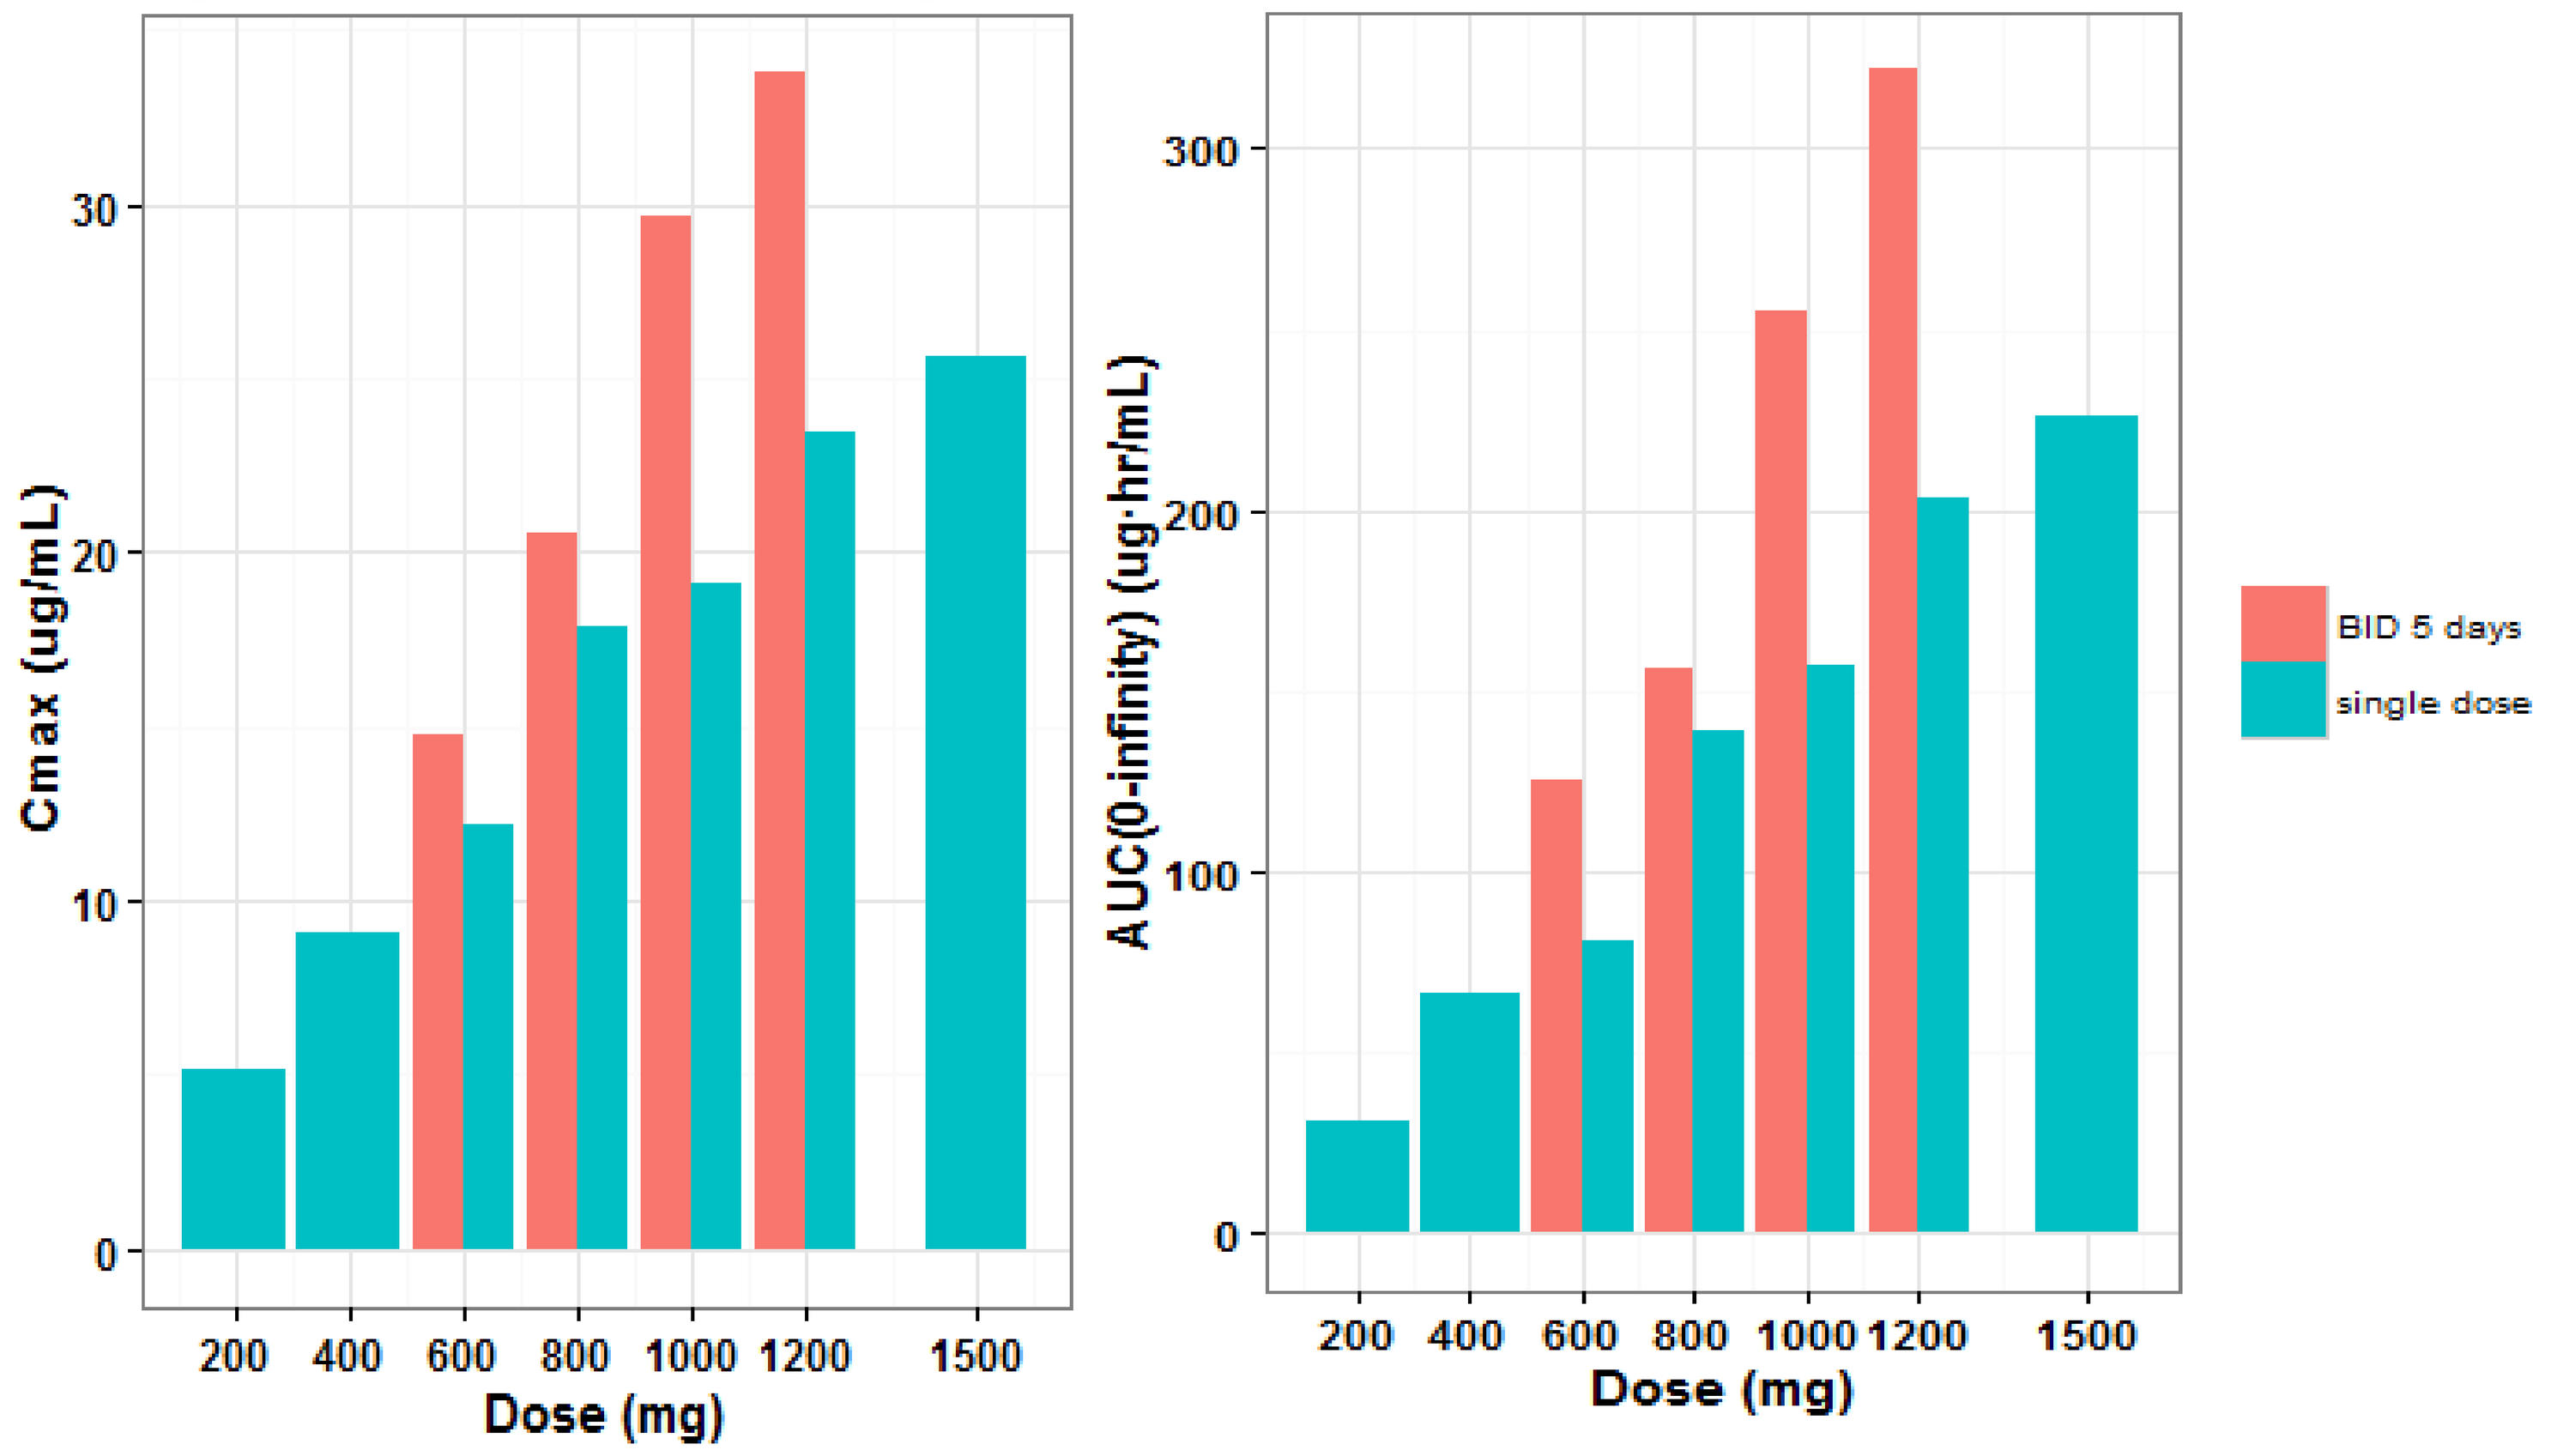
Cmax (ug/mL)
Dose (mg)
AUC(0-infinity) (ug·hr/mL)
Dose (mg)
BID 5 days
single dose

- Linear increase in C_{max} and $AUC_{(0\text{-}infinity)}$ was observed from 200 – 1500 mg single doses.
- For BID for 5 days, the $AUC_{(tau)}$ at steady state was not significantly different from the $AUC_{(0\text{-}infinity)}$ after respective single doses, and the accumulation was minimal.

Conclusions

Conclusions: Single and multiple dose PK studies for WCK 2349 show that the drug was well tolerated and the PK are linear across all dose levels.

References

1.Cercenado, E. & Ruiz de Gopegui, E. [Community-acquired methicillin-resistant Staphylococcus aureus]. *Enfermedades Infecc. Microbiol. Clínica* **26 Suppl 13,** 19–24 (2008).
2.Harris, S. R. *et al.* Evolution of MRSA during hospital transmission and intercontinental spread. *Science* **327,** 469–474 (2010).